iNBHC 2nd International Neurodegeneration and Brain Health Conference
September 25-27, 2026
Hilton Belgrade Hotel, Serbia

Early Versus Late Ocrelizumab Initiation in Relapsing-Remitting Multiple Sclerosis: A Retrospective, Multicenter Cohort Study

Ebru Hatun ULUDAŞDEMİR [a], İpek Güngör DOĞAN [a], Damla ÇETİNKAYA TEZER [a], Melih TÜTÜNCÜ [b], Elif Hilal VURAL [c], Serkan DEMİR [a]

[a] University of Health Sciences, Sancaktepe Şehit Prof. Dr. İlhan Varank Training and Research Hospital, Istanbul, Türkiye, [b] Istanbul University-Cerrahpaşa, Cerrahpaşa Faculty of Medicine, Istanbul, Türkiye, [c] University of Lokman Hekim. Medical Pharmacology Department, Ankara, Turkiye.

Funding: This study was sponsored by F. Hoffmann-La Roche Ltd.

Objectives: Ocrelizumab, a highly effective disease-modifying therapy (HET-DMT), is a B-cell–depleting anti-CD20 monoclonal antibody used in the treatment of relapsing forms of multiple sclerosis (MS) and primary progressive MS.

This study aimed to investigate the long-term clinical, economic, and broader societal outcomes of initiating Ocrelizumab as first-line therapy in patients with relapsing-remitting multiple sclerosis (RRMS), compared with switching to Ocrelizumab after at least one year of treatment with a moderate-efficacy DMT, including its potential impact on productivity, functional independence, and the broader societal burden of MS.

Methods: This active-comparator, retrospective cohort study included patients who were followed at Sancaktepe Şehit Prof. Dr. İlhan Varank Training and Research Hospital and Cerrahpaşa Medical Faculty Hospital between May 2018 and February 2025, had been diagnosed with RRMS according to the 2010/2017 McDonald criteria, and met the predefined inclusion criteria.

Patients were divided into two groups according to the timing of Ocrelizumab initiation: the **Early Initiation Group (EIG)** - First line ocrelizumab group (n=100), comprising patients who received Ocrelizumab directly without prior exposure to any DMT, and the **Late Initiation Group (LIG)** - Escalation to ocrelizumab group (n=100), comprising patients who had received a low-to-moderate-efficacy DMTs (interferon beta 1A, glatiramer acetate, teriflunomide, dimethyl fumarate, fingolimod, cladribine, natalizumab) for at least one year before initiating Ocrelizumab.

Expanded Disability Status Scale (EDSS) scores, Symbol Digit Modalities Test (SDMT), 9-Hole Peg Test (9-HPT), Timed 25-Foot Walk Test (25FWT), relapse history, magnetic resonance imaging (MRI) findings, and treatment-related costs were evaluated.

As Ocrelizumab's public institution discount is confidential, a standard 41% discount was applied to estimate public pharmaceutical costs in the economic analysis.

Results: A total of 200 patients from two centers were included in the study, with 100 patients in the EIG and 100 in the LIG. The groups were similar with respect to age and sex.

The key findings were as follows (see Tables 1–3 and Figure 1 for details):

- The proportion of employed patients was significantly higher in the EIG (p<0.001).
- The time from diagnosis to initiation of the first DMT was significantly longer in the EIG (p<0.001).
- EDSS scores at diagnosis were significantly higher in the EIG (p<0.001).
- At the end of follow-up, the mean EDSS score remained unchanged in the EIG, whereas it increased significantly in the LIG (2.60±1.26 vs 2.92±1.02, p=0.028).
- Despite initiating therapy at a worse baseline EDSS score, patients in the EIG maintained a stable mean EDSS.
- The EIG had significantly higher baseline SDMT scores and shorter 25FWT times.
- Earlier Ocrelizumab use was associated with fewer relapses and markedly lower relapse-related costs (TRY 63.759,14 vs TRY 1.415.129,24).
- On a per patient-year basis, the Ocrelizumab drug cost was 399.261,28 TL in the EIG and 425.684,65 TL in the LIG. In the LIG, an additional first-treatment drug cost of 126.816,54 TL per patient-year was incurred.
- Total productivity loss due to the disease was 298.771,36 TL in the EIG compared with 1.914.238,64 TL in the LIG.

Table 1. Demographic characteristics of the patients at study baseline.

Variable		Early initiation N=100			Late initiation N=100	p
		Mean±SD	Median	Mean±SD	Median	
Age (years)		37.2 ± 10.6	37.1 (IQR 16.7)	38.6 ± 8.5	38.1 (IQR 12.4)	0.197*
Age at diagnosis (years)		32.3 ± 10.4	31.4 (IQR 16.6)	30.3 ± 8.3	29.8 (IQR 13.9)	0.265*
Time from diagnosis to last Ocrelizumab cycle- Total follow-up period (years)		4.99 ± 4.85	3.67 (IQR 4.73)	8.39 ± 4.75	7.38 (IQR 5.36)	<0.001*
Time from diagnosis to first DMT (years)		3.13 ± 4.83	0.74 (IQR 4.22)	0.25 ± 0.85	0.00 (IQR 0.00)	<0.001*
Ocrelizumab usage period (years)		1.98 ± 1.46	1.96 (IQR 2.12)	2.14 ± 1.46	1.69 (IQR 1.54)	0,570*
Sex (female/male, %)		65/35		68/32		0.653**
Employment status (no/yes, %)		5/95		36/64		<0.001*
Frequency of hospital visits (%)	Every 3 months	17		15		1.000‡
	Every 3–6 months	80		84		
	Every 6–9 months	3		1		

* Mann–Whitney U test; ** Pearson's chi-square test, ‡Mantel-Haenzsel test

Table 2. MS-related clinical and functional assessment data.

Variable		Early initiation N=100		Late initiation N=100		p	
		Mean±SD	Median	Mean±SD	Median		
Baseline EDSS		2.60 ± 1.25	2.25 (IQR 1.50)	1.49 ± 0.60	1.00 (IQR 1.00)	<0.001*	Early initiation 1.000‡
Ocrelizumab initial EDSS score		2.60 ±1.25	2.25 (IQR 1.50)	2.85 ±1.01	2.50 (IQR 1.50)	0.078	
EDSS at end of treatment		2.60 ± 1.26	2.50 (IQR 1.38)	2.92 ± 1.02	3.00 (IQR 1.50)	0.028*	Late initiation <0.001‡
Change in EDSS		0.00 ± 0.16	0.00 (IQR 0.00)	1.44 ± 0.97	1.00 (IQR 1.00)	<0.001*	
Baseline SDMT		61.18 ± 7.31	62.00 (IQR 6.75)	53.08 ± 16.22	59.50 (IQR 19.75)	<0.001*	
Baseline 9HPT, right hand		23.47 ± 5.29	22.00 (IQR 2.65)	24.07 ± 4.29	22.74 (IQR 4.00)	0.074*	
Baseline 9HPT, left hand		24.94 ± 4.37	23.90 (IQR 2.95)	25.68 ± 4.60	24.43 (IQR 5.47)	0.204*	
Baseline 25FWT		5.37 ± 1.38	5.00 (IQR 1.15)	5.85 ± 2.10	5.20 (IQR 1.82)	0.020*	
Baseline EDSS distribution (n, %)	0–2.5	59 (59%)		96 (96%)		<0.001**	
	3–5.5	40 (40%)		4 (4%)			
	6–7.5	1 (1%)		0 (0%)			
Proportion of patients experiencing relapse during treatment (n, %)		12 (12%)		75 (75%)		<0.001**	

* Mann–Whitney U test; ** Mantel–Haenszel test; ‡ Wilcoxon test (within-group repeated-measures analysis).

Table 3. Treatment and follow-up costs by treatment period.

Cost item (TL)	Early initiation N=100	Late initiation N=100	Cost per patient-year, EIG	Cost per patient-year, LIG (288.08 patient-years)
First treatment drug cost		36.533.731,20		126.816,54
First treatment outpatient/follow-up cost		244.145,46		847,48
First treatment testing/follow-up cost		37.615,33		130,57
First treatment imaging cost		252.381,45		876,07
First treatment relapse-related costs*		547.800,37		
First first treatment drug and follow-up total cost		37.615.673,81		
			Cost per patient-year, EIG (486 patient-years)	Cost per patient-year, LIG (232.33 patient-years)
Ocrelizumab drug cost	194.040.983,69	98.900.733,93	399.261,28	425.684,65
Ocrelizumab testing/follow-up cost	155.279,43	81.307,26	319,51	349,96
Ocrelizumab imaging cost	412.273,41	217.943,30	848,30	938,06
Ocrelizumab outpatient/follow-up cost	634.434,50	299.532,00	1.305,42	1.289,23
Ocrelizumab relapse-related costs*	63.759,14	867.328,87		
Ocrelizumab treatment drug and follow-up total cost	195.306.730,17	100.366.845,36		
Total productivity loss	**298.771,36**	**1.914.238,64**		
Transportation cost per month	**356.500,00**	**258.000,00**		

*Cost of pulse steroid therapy, plasmapheresis, post-attack hospitalization, and physical therapy

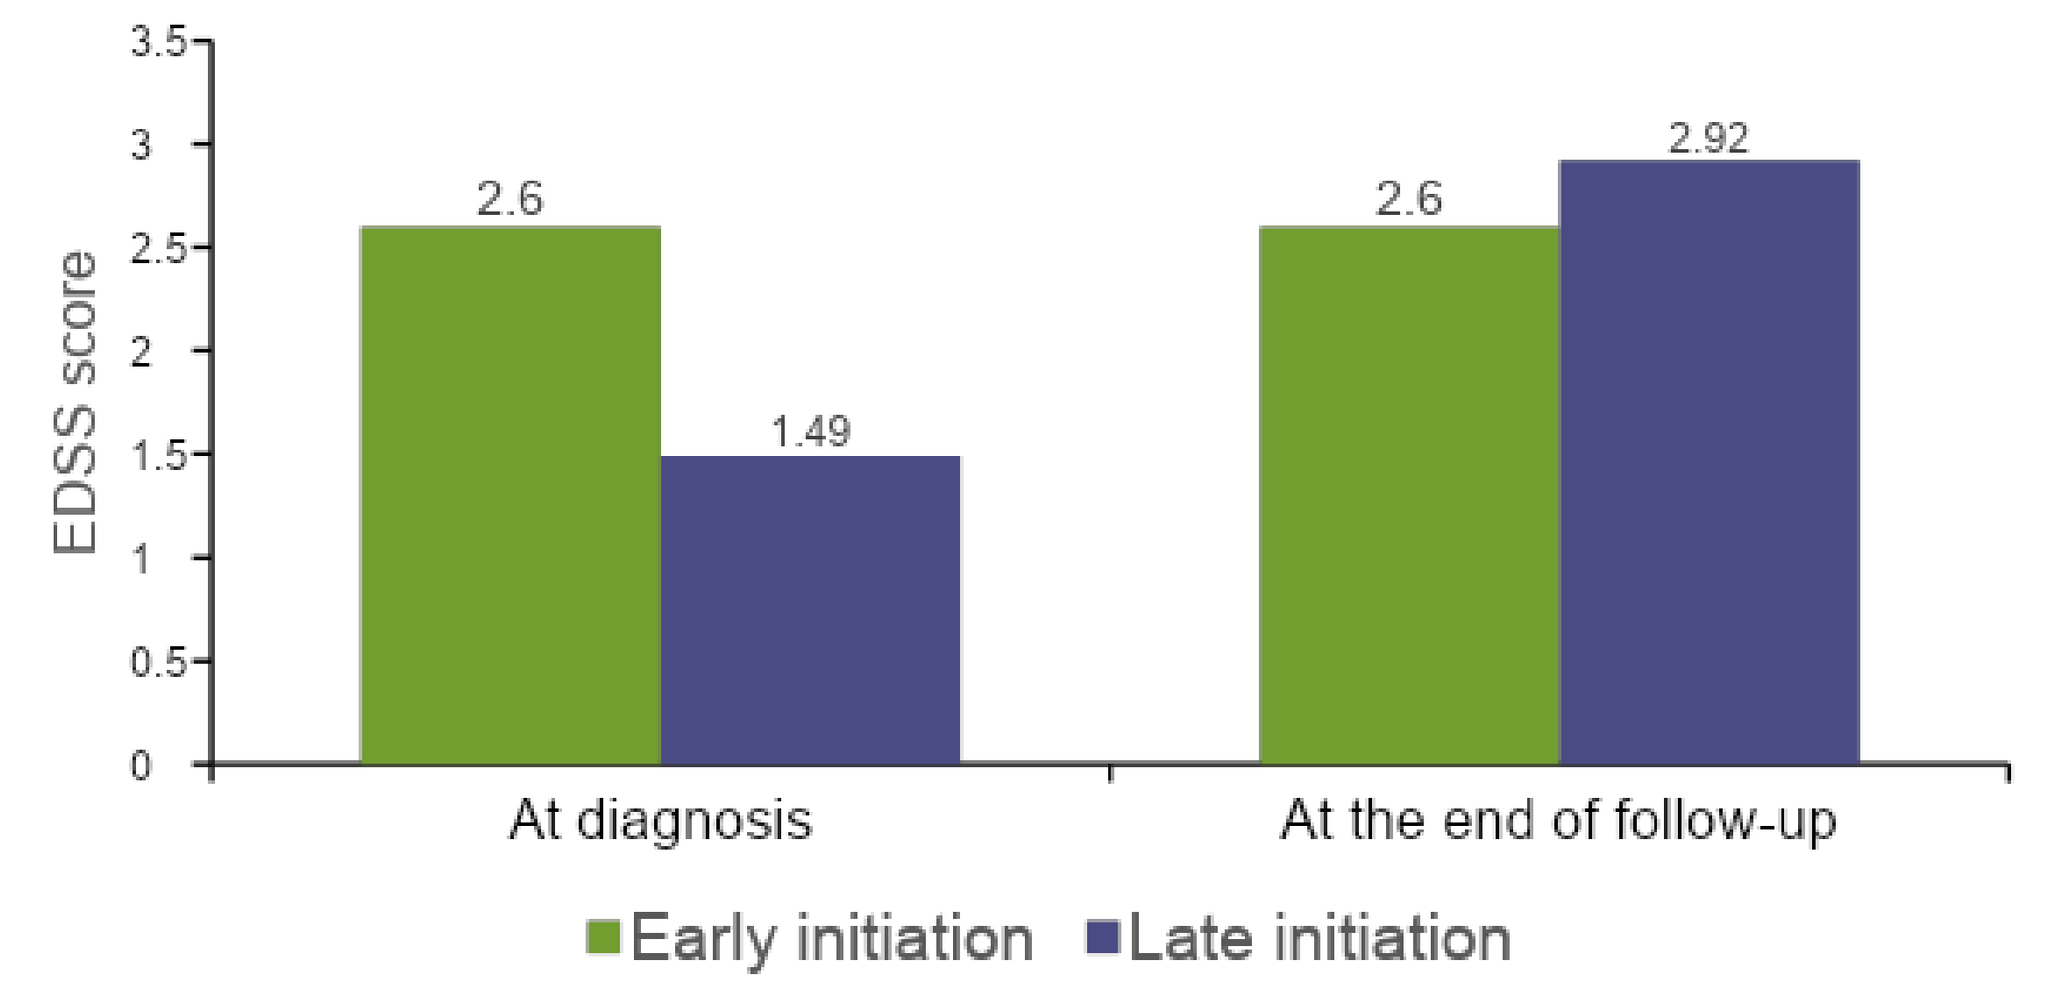
3.5
3
2.5
2
1.5
1
0.5
0
EDSS score
2.6
1.49
2.6
2.92
At diagnosis
At the end of follow-up
Early initiation
Late initiation

Figure 1. Change in EDSS in the two treatment groups.

Discussion: The baseline findings of this study indicate that the Early and LIGs were not completely balanced with respect to disease burden and disability at baseline. Despite having a longer time from diagnosis to first DMT and higher baseline EDSS scores, patients in the EIG had better SDMT performance.This finding suggests that the groups reflected distinct disease trajectories rather than simply "milder" versus "more severe" disease, as EDSS remained stable in the EIG but increased significantly over time in the LIG despite lower disability at diagnosis.

Similarly, MRI findings differed between the groups, with spinal cord/infratentorial lesions being more prominent in the EIG, while the LIG more frequently showed combinations of high T2 lesion burden and other MRI activity features.

When clinical outcomes during treatment were evaluated, the absence of a significant change in EDSS in the EIG, compared with the significant increase observed in the LIG (p<0.001), suggests that earlier initiation of Ocrelizumab is associated with reduced disability progression in patients with RRMS.

These findings may also be considered in the context of the relationship between MRI characteristics and clinical impairment. In MS, spinal cord involvement is known to be particularly associated with physical disability. Spinal cord imaging findings have been shown to correlate with EDSS scores, walking ability, and manual dexterity, while spinal cord atrophy and lesion burden have been reported to make important contributions to the degree of physical disability (Lukas 2013; Azzimonti 2025). Cognitive impairment, in contrast, has been emphasized to be more closely associated with supratentorial lesion burden, cortical lesions, and pathology affecting frontal, parietal, and temporal regions and their associated networks (Wybrecht 2017).

From this perspective, cognitive changes, like physical impairment, may substantially affect activities of daily living, interpersonal relationships, quality of life, and occupational functioning in patients with MS. Therefore, early recognition, assessment, and appropriate management of cognitive impairment are important components of comprehensive MS care.

Another noteworthy finding regarding clinical outcomes was the marked difference in proportion of patients experiencing relapse between the groups during treatment (LIG 75% vs. EIG 12%, p<0.001). This significant difference suggests that the use of Ocrelizumab at an earlier stage of treatment is associated with lower disease activity.

From a societal value perspective, the late initiation strategy resulted in 1.076 additional sick leave days and an additional productivity loss of approximately 1.62 million TRY.

Overall, although the two groups had different clinical and radiological characteristics at baseline, patients in the EIG demonstrated EDSS stability and a markedly lower proportion of patients experiencing relapse during follow-up. These findings support the clinical benefit of initiating Ocrelizumab earlier in the treatment course of appropriately selected patients with RRMS.

The economic findings suggest that the timing of ocrelizumab initiation may have important implications beyond direct treatment costs. Although the overall treatment and follow-up cost was higher in the EIG, the ocrelizumab drug cost per patient-year was lower (399.261,28 vs. 425.684,65 TL), while disease-related productivity loss was markedly lower (298.77,.36 vs. 1.914.238,64 TL). These findings highlight the importance of considering the broader societal burden when evaluating treatment strategies and suggest that earlier initiation of highly effective therapy may provide potential economic and societal benefits. We believe that the economic impact of treatment strategies should be assessed not only in terms of direct healthcare costs, but also encompassing societal consequences such as loss of productivity.

References

*Azzimonti M, Preziosa P, Pagani E, Meani A, Margoni M, Rubin M, Gueye M, Esposito F, Filippi M, Rocca MA. Cervical spinal cord gray matter damage predicts disability worsening in multiple sclerosis: a longitudinal study. J Neurol. 2025 Feb 25;272(3):228. doi: 10.1007/s00415-025-12979-4. PMID: 39998642. *Lukas C, Sombekke MH, Bellenberg B, Hahn HK, Popescu V, Bendfeldt K, Radue EW, Gass A, Borgwardt SJ, Kappos L, Naegelin Y, Knol DL, Polman CH, Geurts JJ, Barkhof F, Vrenken H. Relevance of spinal cord abnormalities to clinical disability in multiple sclerosis: MR imaging findings in a large cohort of patients. Radiology. 2013 Nov;269(2):542-52. doi: 10.1148/radiology.13122566. Epub 2013 Jun 4. PMID: 23737540. *Wybrecht D, Reuter F, Pariollaud F, Zaaraoui W, Le Troter A, Rico A, Confort-Gouny S, Soulier E, Guye M, Maarouf A, Ranjeva JP, Pelletier J, Audoin B. New brain lesions with no impact on physical disability can impact cognition in early multiple sclerosis: A ten-year longitudinal study. PLoS One. 2017 Nov 17;12(11):e0184650. doi: 10.1371/journal.pone.0184650. PMID: 29149177; PMCID: PMC5693435

inbhc2026.org